Iron absorption is maintained over time following repeated twice-daily ferric maltol dosing: *Post hoc* analysis of a multicenter, open-label, randomized, Phase 1 clinical study

Stefanie Howaldt, MD[1], Michael L. Cody, MD[2], Jacqueline A. Mitchell, MA, DPhil (Oxon)[3]

[1]HaFCED e.K., Hamburg, Germany; [2]Shield Therapeutics, Dallas, TX, USA; [3]Shield Therapeutics, Newcastle upon Tyne, UK

Scan the QR codes to access a copy of this poster and other relevant publications on ferric maltol at www.shieldtherapeutics.com/products/publications/. Copies of this poster and other documents are for personal use only and may not be reproduced without written permission from the authors or publishers.

LEARNING OBJECTIVES

This poster will help you to:

- Explain how iron deficiency affects patients
- Describe the impact of repeated twice-daily ferric maltol dosing on iron uptake in patients with iron deficiency
- Discuss the benefits of ferric maltol in patients with iron deficiency

INTRODUCTION

- The amount of iron in the body is normally tightly regulated (**Figure 1**); however, these regulatory processes can be disturbed by many conditions, such as malnutrition, malabsorption, blood loss, or inflammation, resulting in iron deficiency[1]
- Iron deficiency is the most common nutritional disorder in the world, accounting for almost two thirds of the estimated 1.92 billion cases of anemia worldwide, and it is particularly common in women of reproductive age[2]
- Iron deficiency can result in fatigue, headache, restless legs, and other physical and cognitive impairments, severely limiting daily activities and quality of life[3–5]
- Patients with iron deficiency may require long-term iron replacement therapy to replenish iron stores. However, traditional oral iron salts are often associated with poor absorption and intolerable gastrointestinal toxicity, due to unabsorbed iron and the generation of reactive oxygen species[3,6,7]
- Ferric maltol is an oral iron formulation that was uniquely designed to improve gastrointestinal absorption, resulting in increased iron uptake while minimizing the risk of intestinal damage and disruption to the gut microbiota[8,9]

Figure 1. Physiologic iron regulatory pathways

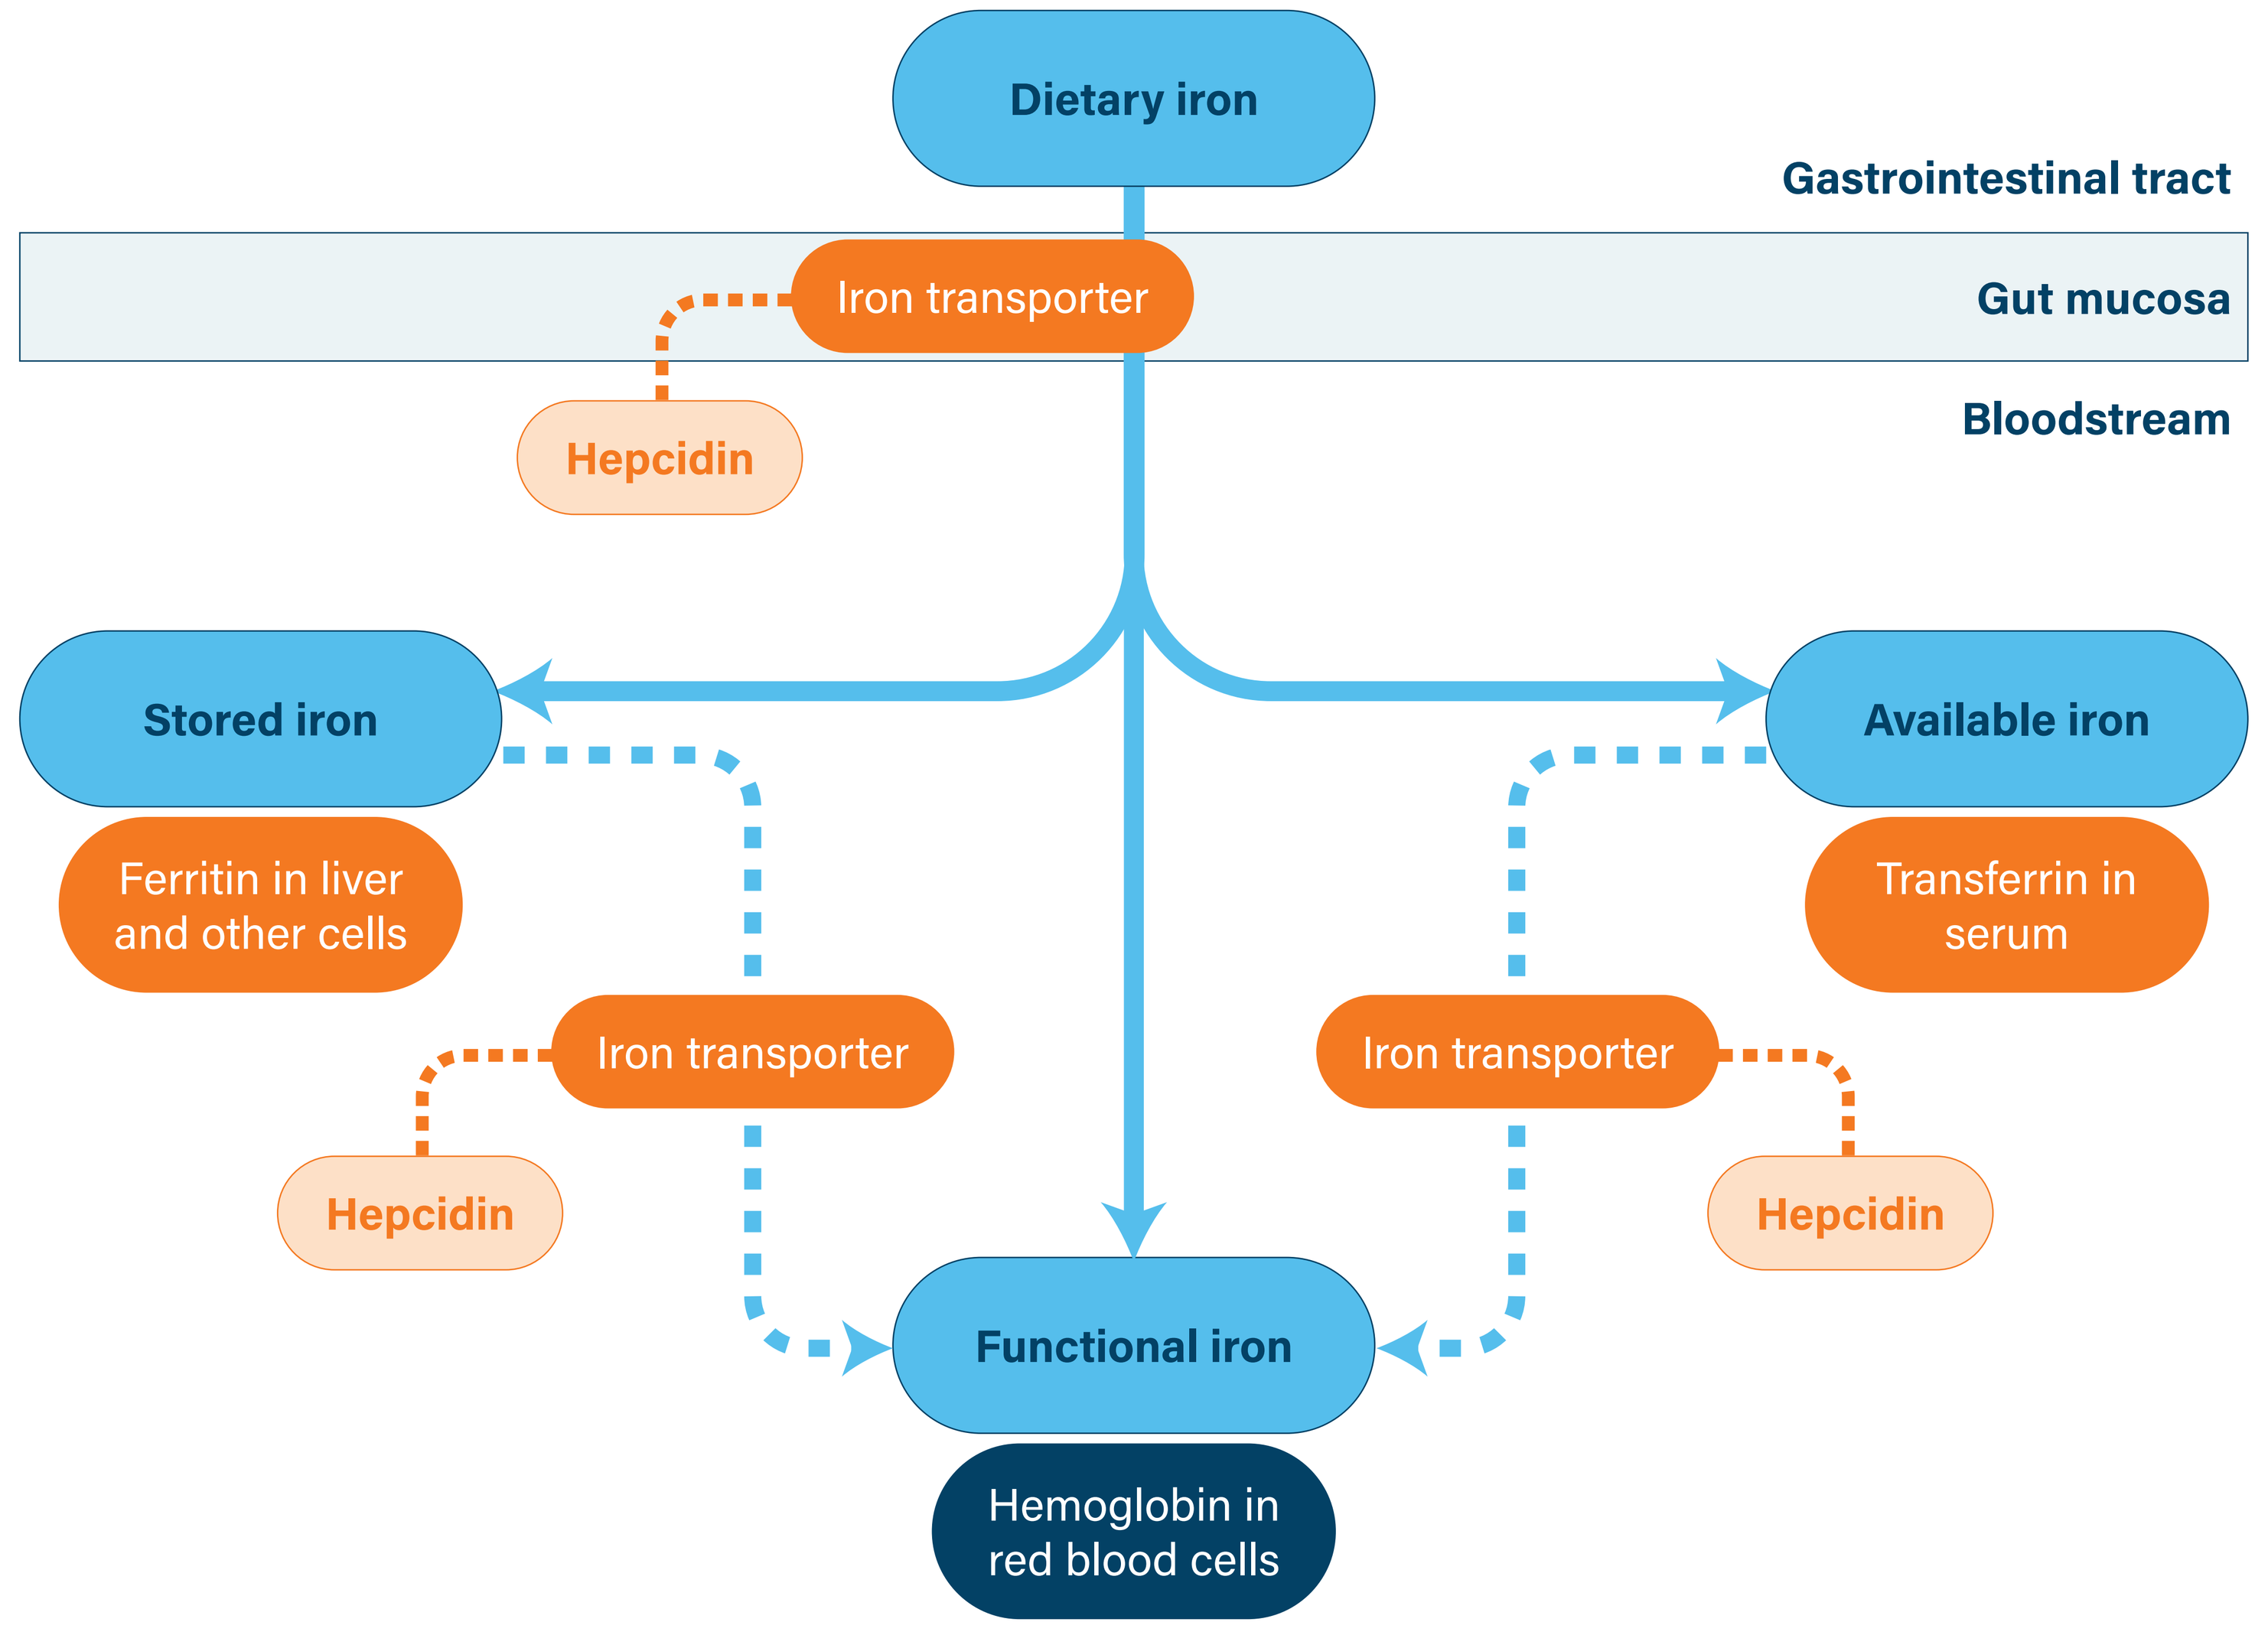

Dietary iron
Gastrointestinal tract
Iron transporter
Gut mucosa
Hepcidin
Bloodstream
Stored iron
Available iron
Ferritin in liver and other cells
Transferrin in serum
Iron transporter
Iron transporter
Hepcidin
Hepcidin
Functional iron
Hemoglobin in red blood cells

PURPOSE

- Because the body tightly regulates iron absorption to prevent iron overload, we assessed whether iron uptake is restricted following repeated ferric maltol dosing

METHODS

- Study design (**Figure 2**)
 - *Post hoc* analysis of a German multicenter Phase 1 study[10]
- Study participants
 - Adults with inflammatory bowel disease and iron deficiency
- Study treatment
 - Ferric maltol 30 mg or 60 mg orally twice daily for 7 days followed by a single dose on the morning of Day 8
- Measures
 - Total serum iron and TSAT following the morning dose on Days 1 and 8, based on blood samples taken at the time of ferric maltol dosing (= 0 hours) and 0.25, 0.5, 0.75, 1, 1.5, 2, 3, 4, and 6 hours after dosing
- Statistical analysis
 - Data are reported descriptively as mean (standard deviation) C_{max} and AUC_{6h}

Ethical conduct

- The study was conducted in accordance with the principles of the Declaration of Helsinki and the International Council for Harmonisation Good Clinical Practice guideline
- The protocol was approved by state ethics review boards (Ethik-Kommissionen) for the participating study centers: Ethik-Kommission des Landes Sachsen-Anhalt; Ethik-Kommission des Landes Berlin Landesamt für Gesundheit und Soziales; Ethik-Kommission der Ärztekammer Hamburg; Ethik-Kommission der Ärztekammer Westfalen-Lippe und der Medizinischen Fakultät der WWU; Ethik-Kommission bei der Ärztekammer Niedersachsen
- All study participants provided written informed consent prior to participation

Figure 2. Study design

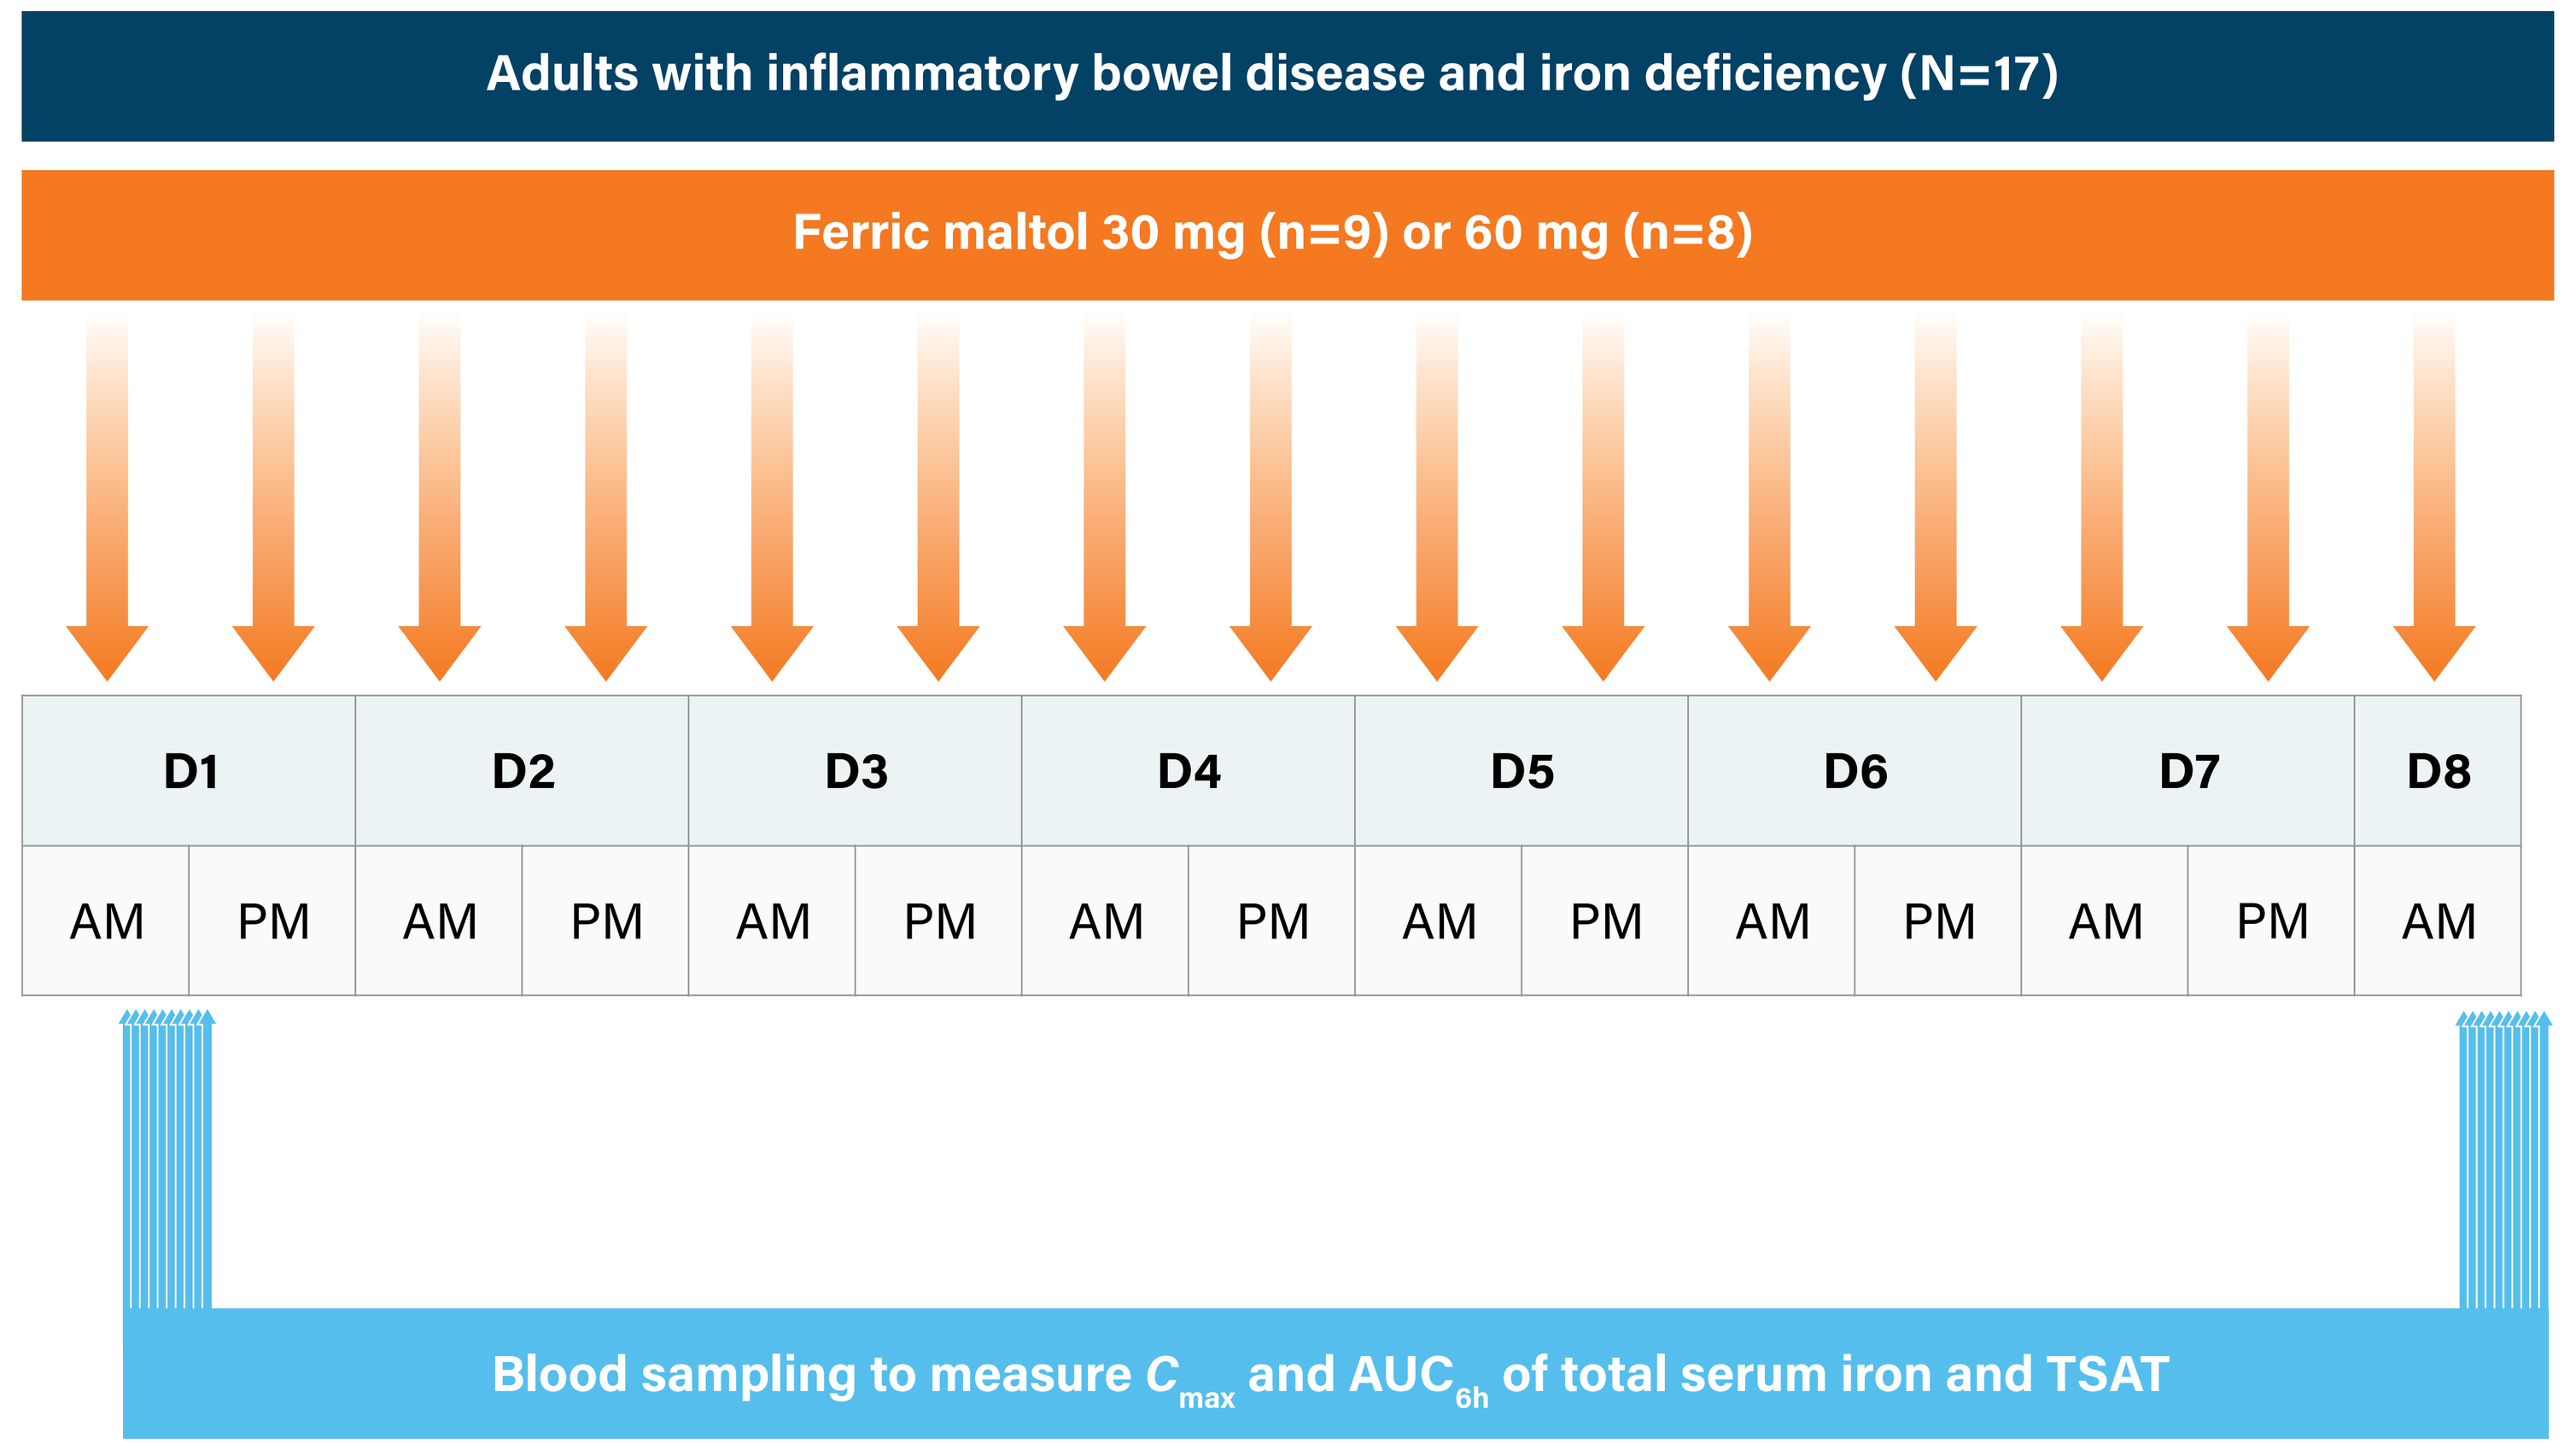

Adults with inflammatory bowel disease and iron deficiency (N=17)
Ferric maltol 30 mg (n=9) or 60 mg (n=8)
D1 D2 D3 D4 D5 D6 D7 D8
AM PM AM PM AM PM AM PM AM PM AM PM AM PM AM
Blood sampling to measure C_{max} and AUC_{6h} of total serum iron and TSAT

AM, morning; AUC_{6h}, area under the curve up to 6 hours; C_{max}, maximum concentration; D, day; PM, evening; TSAT, transferrin saturation.

RESULTS

- Total serum iron (**Figure 3**) and TSAT (**Figure 4**) were only slightly lower by Day 8 of twice-daily ferric maltol dosing versus the first dose, with overlapping error bars between Days 1 and 8 indicating no clinically meaningful difference in iron uptake over time
- Taken together with evidence that baseline levels of serum hepcidin did not affect iron uptake from ferric maltol,[11] it appears that iron regulatory pathways do not inhibit iron uptake following repeated ferric maltol dosing, while maintaining physiologic control to prevent iron overload

Figure 3. Total serum iron measures following the morning dose on Days 1 and 8 of twice-daily ferric maltol dosing

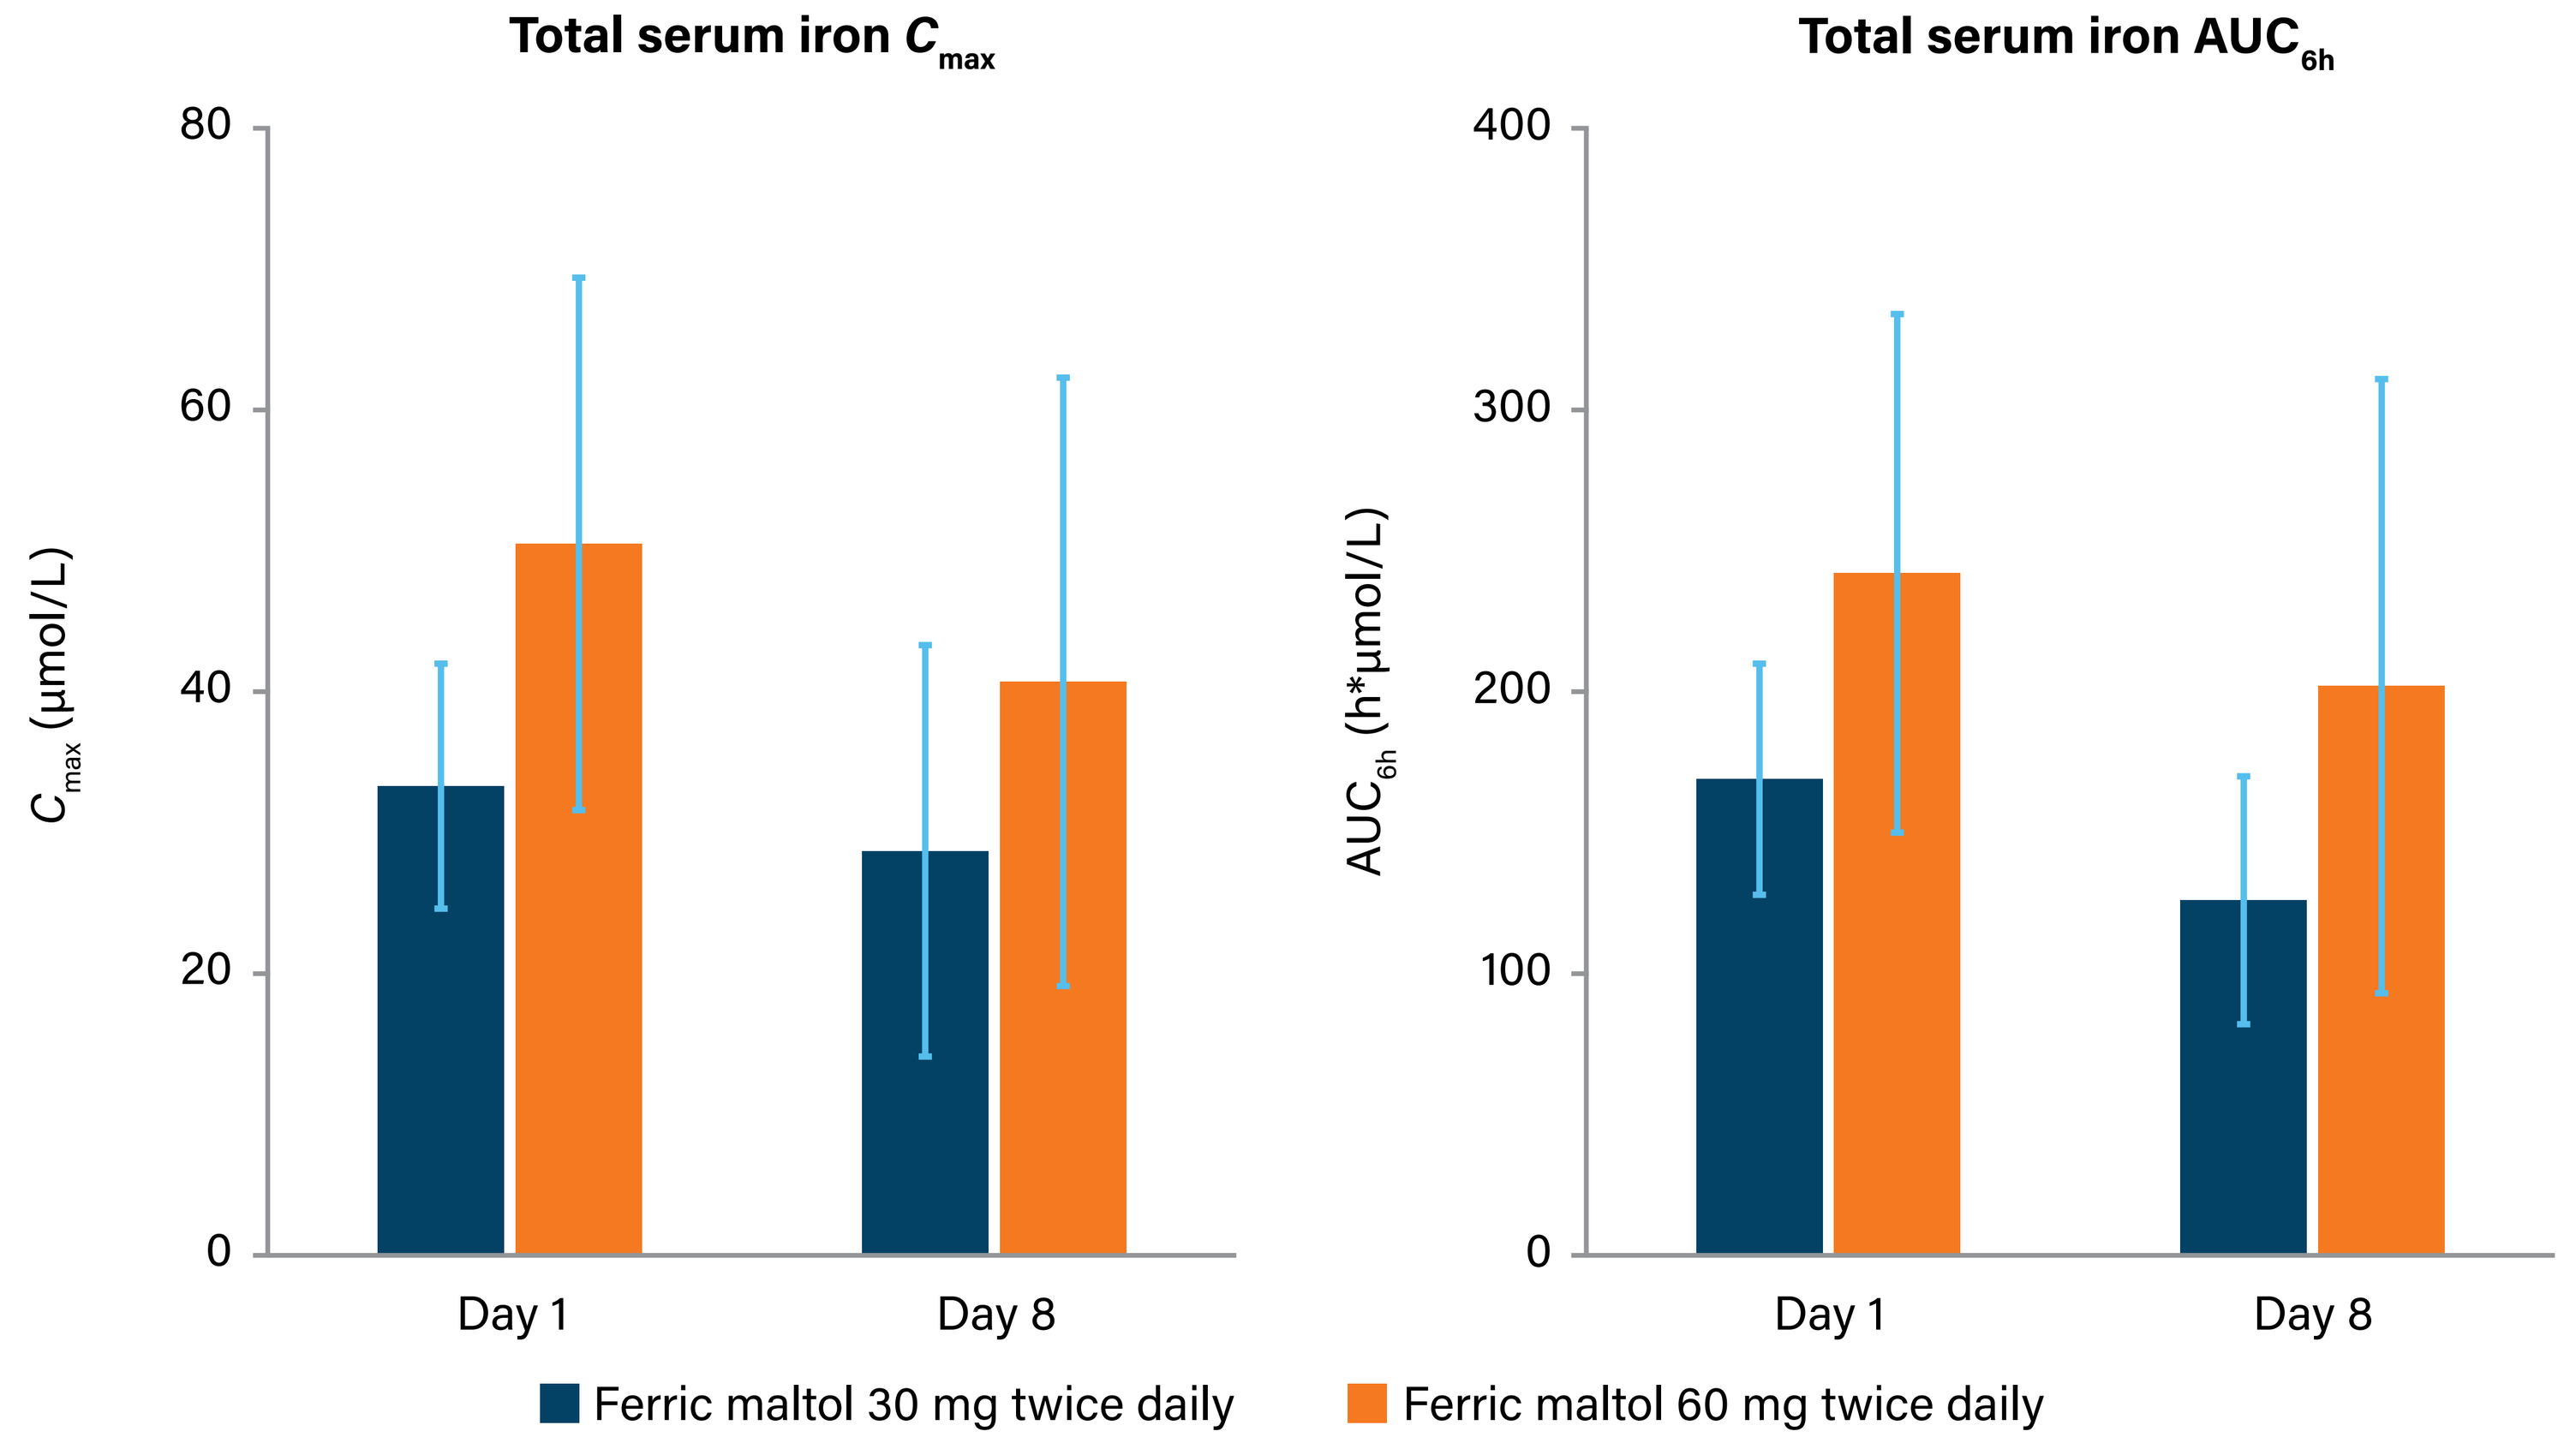

Total serum iron C_{max}
C_{max} (µmol/L)
80
60
40
20
0
Day 1
Day 8
Total serum iron AUC_{6h}
AUC_{6h} (h*µmol/L)
400
300
200
100
0
Day 1
Day 8
Ferric maltol 30 mg twice daily
Ferric maltol 60 mg twice daily

AUC_{6h}, area under the curve up to 6 hours; C_{max}, maximum concentration.

Figure 4. TSAT measures following the morning dose on Days 1 and 8 of twice-daily ferric maltol dosing

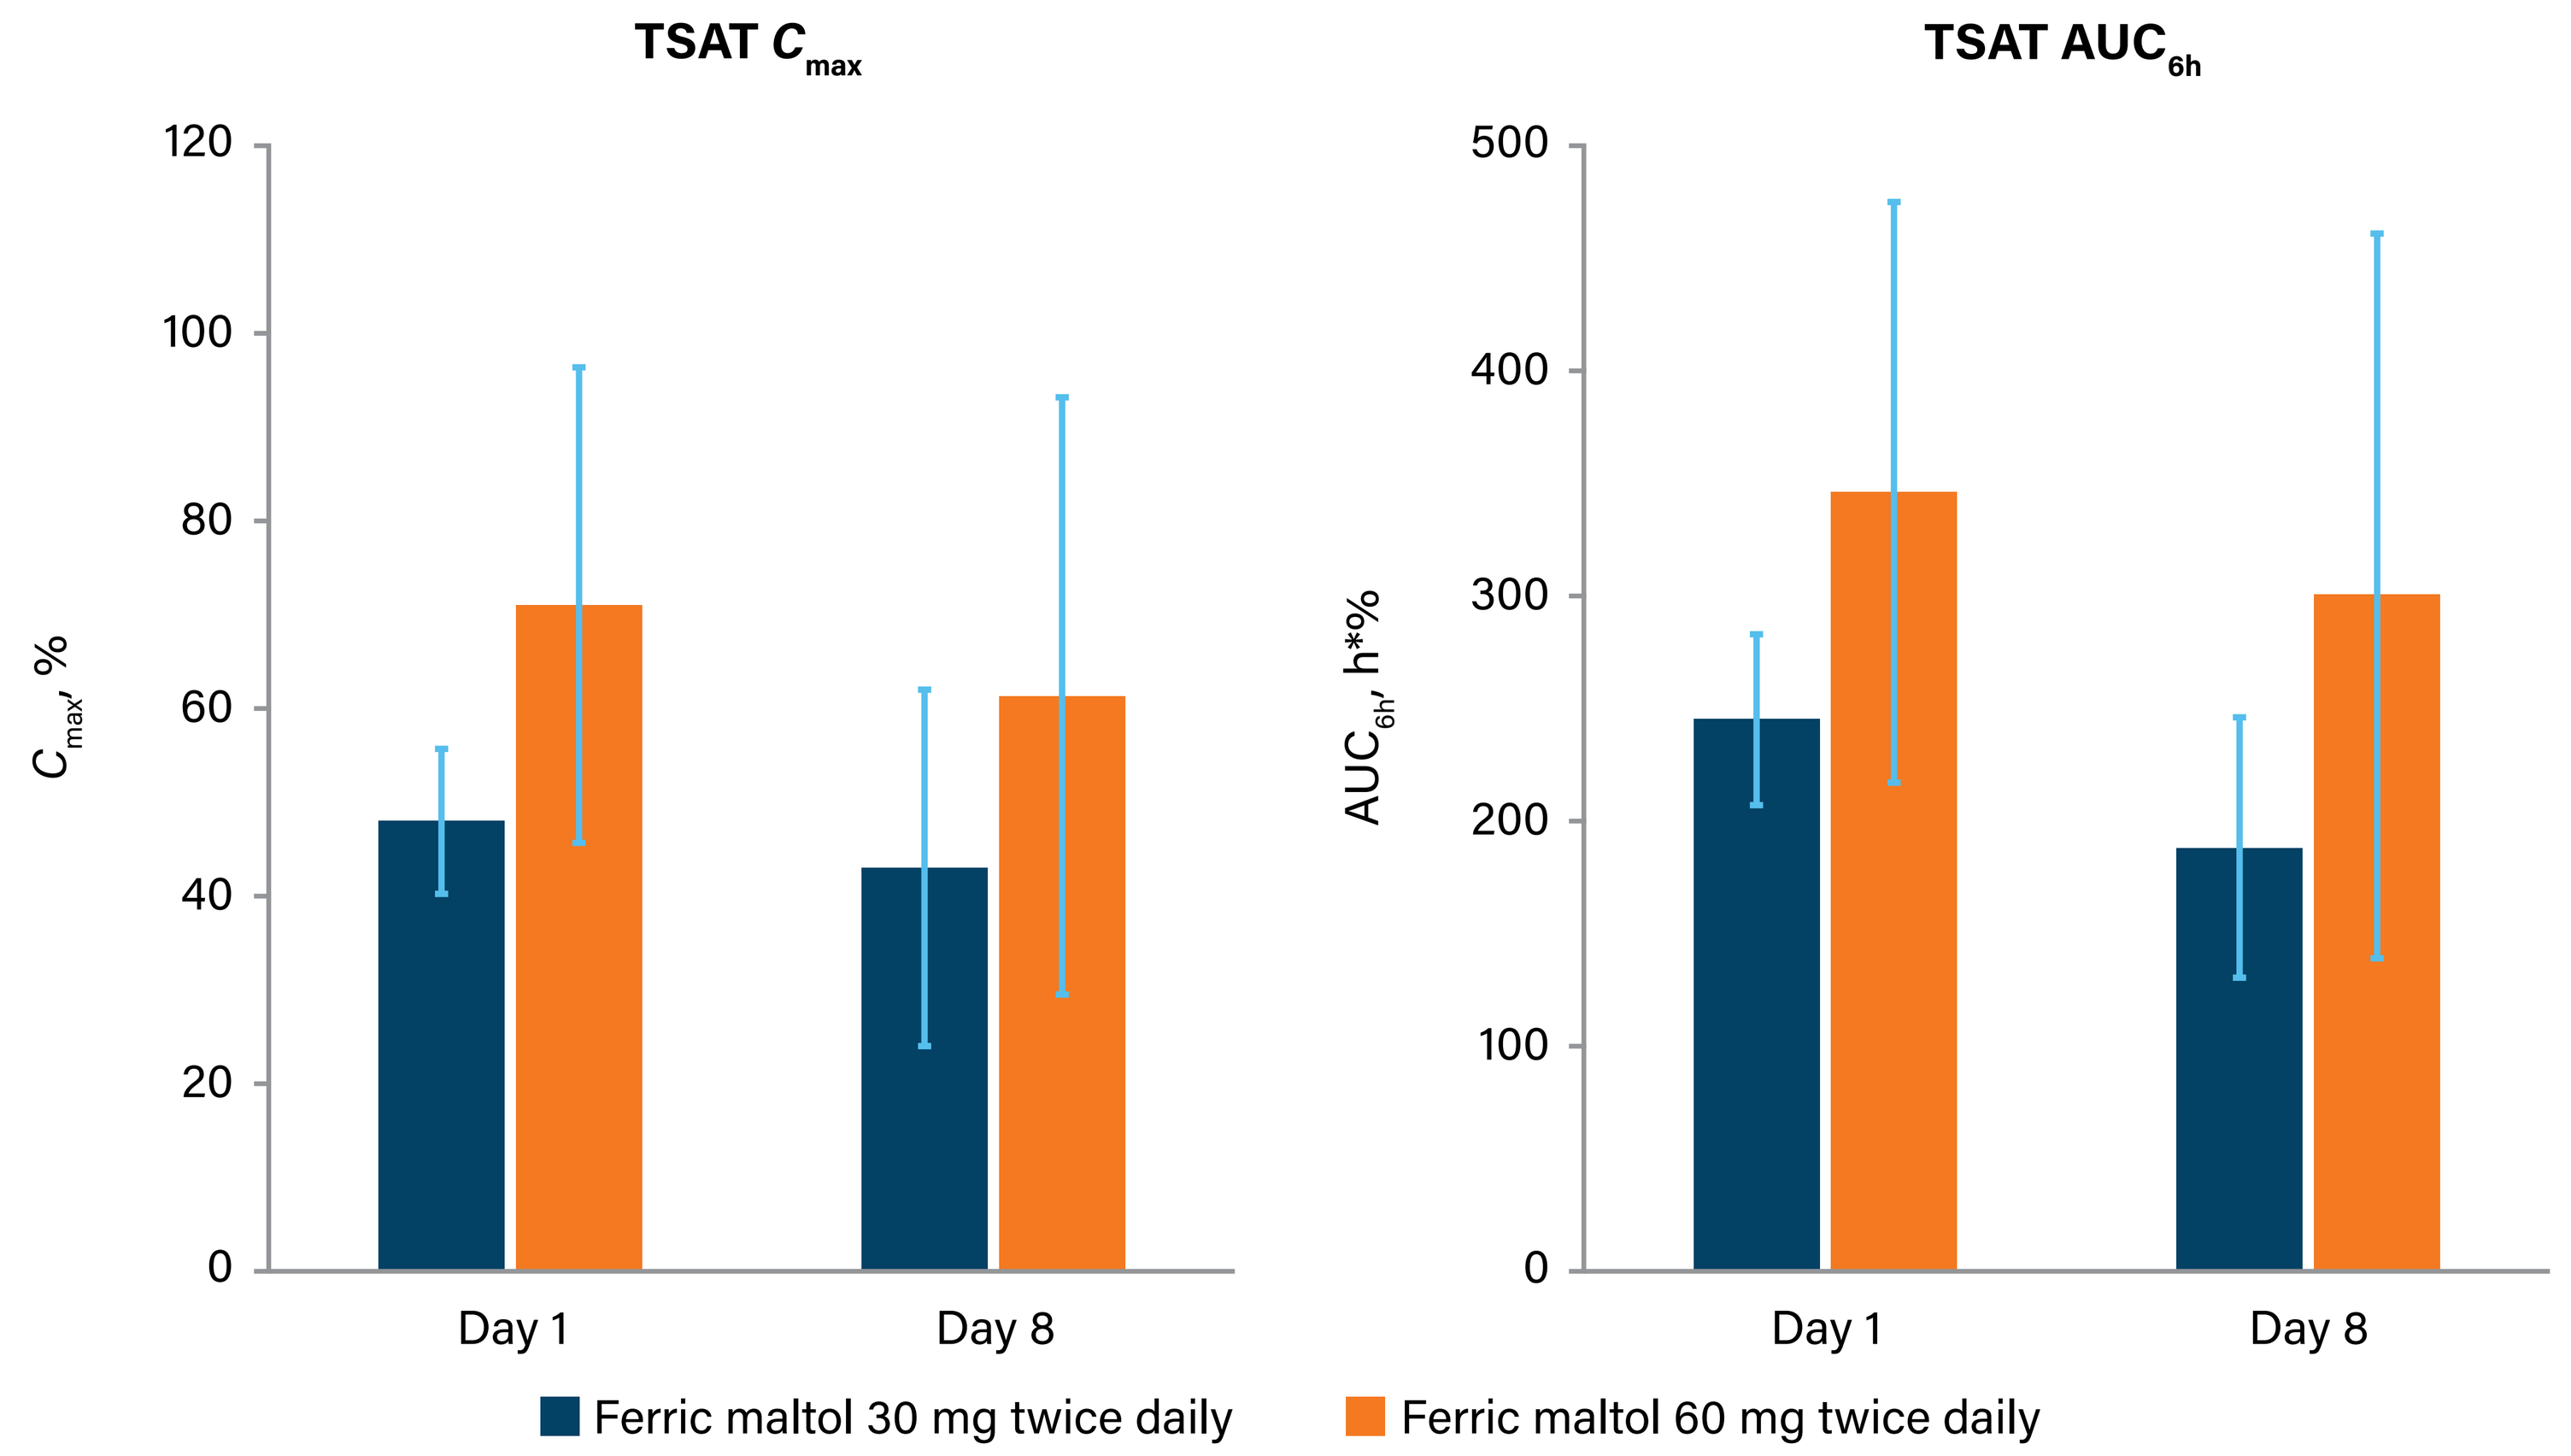

TSAT C_{max}
C_{max}, %
120
100
80
60
40
20
0
Day 1
Day 8
TSAT AUC_{6h}
AUC_{6h}, h*%
500
400
300
200
100
0
Day 1
Day 8
Ferric maltol 30 mg twice daily
Ferric maltol 60 mg twice daily

AUC_{6h}, area under the curve up to 6 hours; C_{max}, maximum concentration; TSAT, transferrin saturation.

CONCLUSIONS

- Iron deficiency is the most common and widespread nutritional disorder leading to anemia in the world[12]
- Iron deficiency affects patients by impairing physical and cognitive function, consequently limiting daily activities and quality of life[3–5]
- Twice-daily ferric maltol dosing improves iron uptake in patients with iron deficiency, with no potentially clinically significant decrease in uptake observed following repeated dosing
- At the approved dose of 30 mg twice daily, ferric maltol can benefit women (and men) with iron deficiency with or without anemia by providing a well-tolerated, effective, and easy-to-take iron replacement therapy option

GLOSSARY

Anemia: any situation where insufficient iron is available to produce red blood cells; anemia is indicated by low hemoglobin levels (<13 g/dL in men, <12 g/dL in nonpregnant women and children aged >11 years, <11 g/dL in pregnant women and children ≤11 years[12])

AUC_{6h} (area under the curve up to 6 hours): the definite integral of the concentration (e.g., of a drug) in blood plasma as a function of time (6 hours); an indicator of the total amount present in the body over a 6-hour period

C_{max} (maximum concentration): the highest concentration (of the substance being measured) in the blood, cerebrospinal fluid, or target organ after a dose is given

Ferric maltol: an oral iron replacement therapy designed to optimize iron absorption in the gut while reducing the gastrointestinal adverse events associated with unabsorbed free iron;[8] it is approved in the treatment of adults with iron deficiency with or without anemia, independent of the underlying condition, and is commercially available in Europe and the United States[9]

Ferritin: a protein that stores iron in cells (**Figure 1**); ferritin production is upregulated in the presence of iron, so low concentrations (e.g., <15 g/L in adults) can indicate iron deficiency. However, ferritin is also upregulated in inflammatory conditions, so a higher cut-off may be required (e.g., <100 g/L) to define iron deficiency in patients with inflammation[1,3,13]

Hemoglobin: a protein in red blood cells that transports oxygen around the body and requires iron for oxygen binding (**Figure 1**); if insufficient iron is available, the body cannot produce hemoglobin or transport oxygen

Hepcidin: a protein produced by the liver that controls the amount of circulating iron by inhibiting iron transport across cells (**Figure 1**); when iron levels are high, hepcidin production is upregulated

Iron deficiency: any situation in which there is insufficient iron available to meet the body's needs. Absolute iron deficiency can develop as a result of malnutrition, malabsorption, or blood loss. Functional iron deficiency occurs when dysfunction in the body's iron regulatory mechanisms prevents iron absorption and release from storage proteins.[1,3,14] Iron deficiency is usually diagnosed using serum levels of the molecules involved in iron regulation, such as ferritin and transferrin, and hemoglobin, with different cut-offs used depending on patient and underlying disease factors

Serum iron: iron that is bound to transferrin and circulating freely in the blood (**Figure 1**); it provides a measure of iron availability

TSAT (transferrin saturation): a measure of the body's actual versus potential capacity for iron uptake. Transferrin is a protein that binds to iron and facilitates iron transporation around the body (**Figure 1**); transferrin molecules have two iron-binding sites, but the average proportion of overall binding sites occupied (i.e., the TSAT) is typically 20–50%. A TSAT of <20% indicates low iron availability and can be used to confirm a diagnosis of iron deficiency, particularly in the presence of low ferritin levels[3,15,16]

REFERENCES

1. Abbaspour N et al. *J Res Med Sci* 2014;19:164–174; **2.** GBD 2021 Anaemia Collaborators. *Lancet Haematol* 2023;10:e713–e734; **3.** Lopez A et al. *Lancet* 2016;387:907–916; **4.** Stein J et al. *Nat Rev Gastroenterol Hepatol* 2010;7:599–610; **5.** Babitt JL, Lin HY. *J Am Soc Nephrol* 2012;23:1631–1634; **6.** Gasche C et al. *Inflamm Bowel Dis* 2015;21:579–588; **7.** Petzer V et al. *Pharmaceuticals (Basel)* 2018;11:135; **8.** Kontoghiorghes GJ et al. *Int J Mol Sci* 2021;22:5546; **9.** Schmidt C et al. *J Clin Med* 2021;10:4448; **10.** Bokemeyer B et al. *Eur J Drug Metab Pharmacokinet* 2017;42:229–238; **11.** Quraishi N et al. *Gut* 2017;66(Suppl 2):A262; **12.** World Health Organization. https://www.who.int/publications/i/item/WHO-NMH-NHD-MNM-11.1; **13.** Dignass AU et al. *J Crohns Colitis* 2015;9:211–222; **14.** Gargallo-Puyuelo CJ et al. *Nutrients* 2018;10:1959; **15.** Beard JL. *J Nutr* 2001;131:568S–580S; **16.** Chua AC et al. *Crit Rev Clin Lab Sci* 2007;44:413–459.

ACKNOWLEDGMENTS

The authors thank the patients, their families, and the investigators involved in the study. Pharmacokinetic analyses were provided by Venn Life Sciences ED BV, Breda, Netherlands, with funding from Shield Therapeutics. Medical writing support was provided by Sara Black, ISMPP CMPP™, of OPEN Health Communications, London, UK, with funding from Shield Therapeutics.

DISCLOSURES

This study was funded by Shield Therapeutics. Stefanie Howaldt has no further conflicts to disclose. Michael L. Cody is currently employed by Shield Therapeutics and holds equity in Shield Therapeutics. Jacqueline A. Mitchell is currently employed by Shield Therapeutics and holds equity in Shield Therapeutics and PharmaKrysto Ltd.

AUTHOR CONTACT

Stefanie Howaldt: howaldt@hafced.de;
Michael L. Cody: michael.cody@shieldtx.com;
Jacqueline A. Mitchell: jackie.mitchell@shieldtx.com

I-2023-108